NUC-3373 induces DAMPs from NSCLC cells potentiating a favorable immunogenic microenvironment

Boback Kaghazchi[1], In Hwa Um[1], Jennifer Bré[1,2], David J. Harrison[1,2], Oliver J. Read[1,2]

1) University of St Andrews, UK
2) NuCana plc, UK

Abstract Number: 128 Email: bk68@st-andrews.ac.uk

BACKGROUND

- Lung 2nd most common cancer ▪ Incidence: 2.2 million ▪ Leading cause of cancer mortality[1]
- NSCLC accounts for ~85% cases of lung cancer, majority are adeno-or squamous carcinoma
 - Systemic treatments include immune checkpoint inhibitors (ICIs), chemotherapy and targeted therapies
- ICIs exert anti-cancer effects by modulating the interaction between tumor and immune cells
- Chemotherapies induce ICD *in vitro*, turning immunologically 'cold' tumors 'hot'[2]

Immune contexture & DAMPs in NSCLC

- Immune contexture refers to the spatial organization and density of immune cells within the tumor microenvironment (TME)[3]
- Increased populations of CD8+ cytotoxic T cells and CD20+ B cells in the TME predict better survival in stage I-III NSCLC[4]
- DAMPs (CRT, HMGB1, ATP) promote immune infiltration within the TME[5]

NUC-3373: A potent TS inhibitor with additional DNA-mediated mode of action

- ProTide transformation of FUDR-MP[6,7], active anti-cancer metabolite of 5-FU
- Resistant to breakdown by DPD
- Low levels of toxic metabolites (FBAL, FUTP)[8,9]
- Generates high levels of FUDR-MP resulting in TS inhibition and DNA damage[9]
- Induces DAMPs in colorectal cancer cells and potentiates ICD[10]

Aims

- Investigate immune contexture in treatment naïve NSCLC patients
- Confirm NUC-3373 induces DAMPs in NSCLC cells leading to immunogenic cell death

Hypotheses

- NUC-3373 induced DAMPs promote a favorable immunogenic environment

NUC-3373: additional modes of action

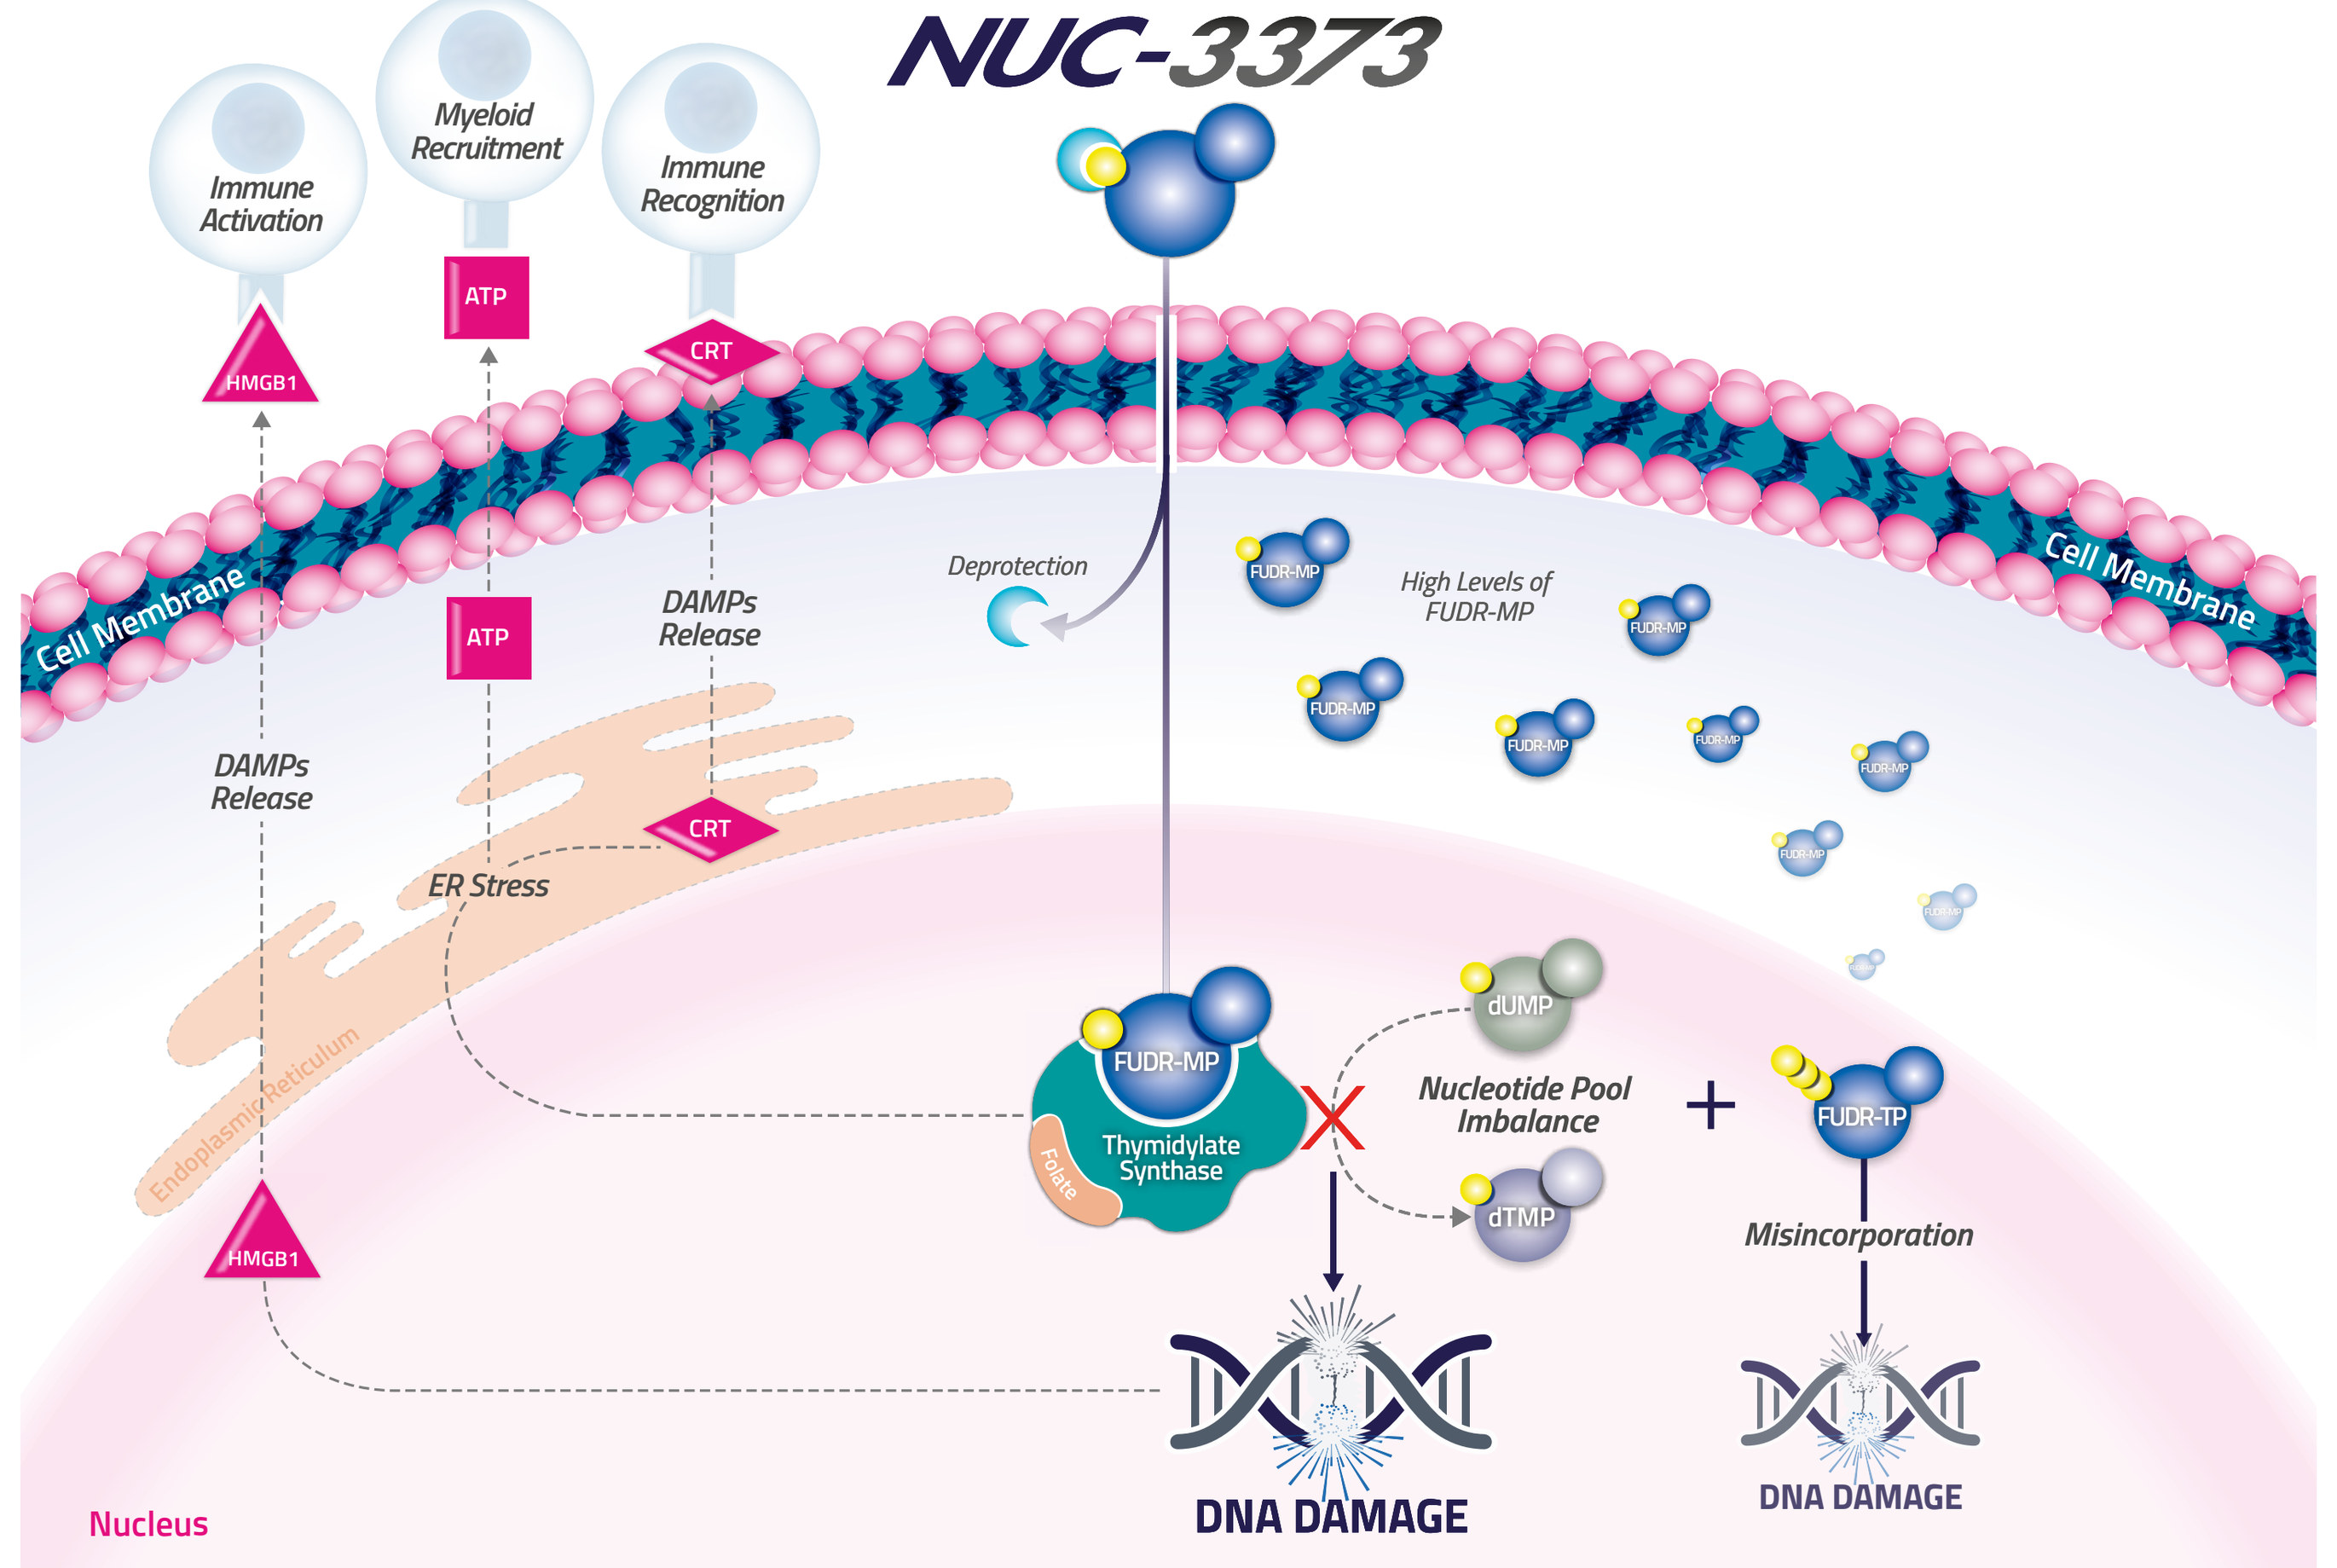

NUC-3373
Immune Activation
Myeloid Recruitment
Immune Recognition
ATP
CRT
HMGB1
Cell Membrane
Deprotection
High Levels of FUDR-MP
FUDR-MP
DAMPs Release
ER Stress
Endoplasmic reticulum
Thymidylate Synthase
Folate
dUMP
dTMP
Nucleotide Pool Imbalance
FUDR-TP
Misincorporation
DNA DAMAGE
Nucleus

Figure 1. In addition to being a potent TS inhibitor with a DNA-mediated mode of action, NUC-3373 can induce ER stress and DAMPs release promoting a favorable immunogenic tumor microenvironment.

METHODS

Survival analysis: Stage I-II lung adenocarcinoma tumors (162 treatment naïve patients) analyzed based on cell morphology and tumor cell populations, stratified according to TME immune cell population (Immune^{High}, n=64, *vs* Immune^{Low}, n=98). Immune^{High} = total immune cells/total cancer cells ≥ 41.7% (determined by outcome-based cut-point optimisation from immune population metrics using 2X-Tiles software).

DAMPs & lymphocyte populations in patient samples: FFPE tissue (sub cohort, n=60) were analyzed for CRT, HMGB1, CD8 and CD20 expression by multiplex immunofluorescence. Infiltration and spatial analyses performed using HALO.

Cell culture: Nx002 (squamous carcinoma) cells were treated with 10 µM NUC-3373 (sub-IC_{50} dose) or vehicle control (DMSO) for 24 hours.

CRT: Cell surface expression assessed by flow cytometry.
ATP: Assessed by quinacrine-mediated fluorescence labelling.
HMGB1: Nuclear expression assessed by immunofluorescence; extracellular release measured by ELISA.
PD-L1: NSCLC cell surface expression assessed by flow cytometry.
ICD: Nx002 cells treated with 10 µM NUC-3373 for 24 hours prior to co-culture with PBMCs. T-cell mediated cell killing ± 10 µg/ml pembrolizumab assessed by automated cytometry analysis (Celigo).
Statistical Analyses: Mantel-Cox Log-rank test was performed for survival analysis (Fig.2).
Wilcoxon test was performed for spatial analyses (Fig.3 & 4).

(for all cell experiments, n=3, unless otherwise stated)

RESULTS

High immune cell populations in TME predict improved survival

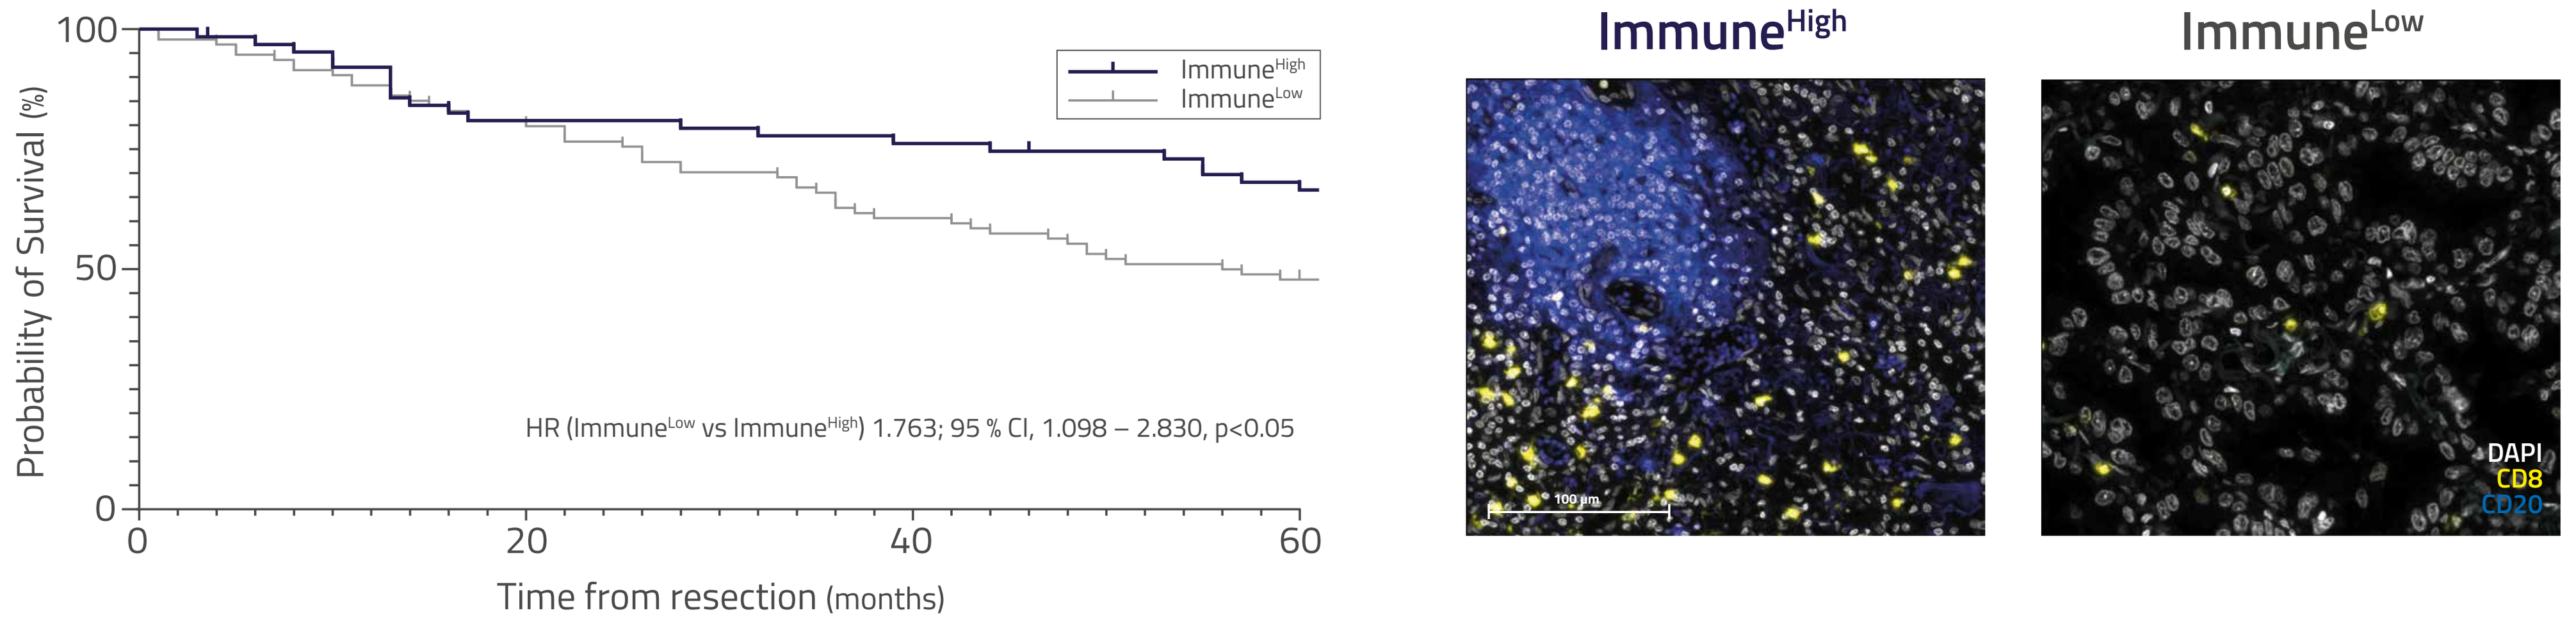

Immune^{High}
Immune^{Low}
Probability of Survival (%)
Time from resection (months)
HR (Immune^{Low} vs Immune^{High}) 1.763; 95 % CI, 1.098 – 2.830, p<0.05
DAPI
CD8
CD20
100 µm

Figure 2. Kaplan-Meier curves for survival of 162 treatment-naïve patients with early-stage lung adenocarcinoma over 5-years. Patients were stratified for TME immune cell population, Immune^{High} n=64; Immune^{Low} n=98.

- High immune cell population within the TME predicts an improved 5-year overall survival
- Immune^{High} *vs* Immune^{Low} 5-year survival = 66% *vs* 48% (p<0.05)

High expression of CRT is associated with increased infiltration of CD8+ cells in TME

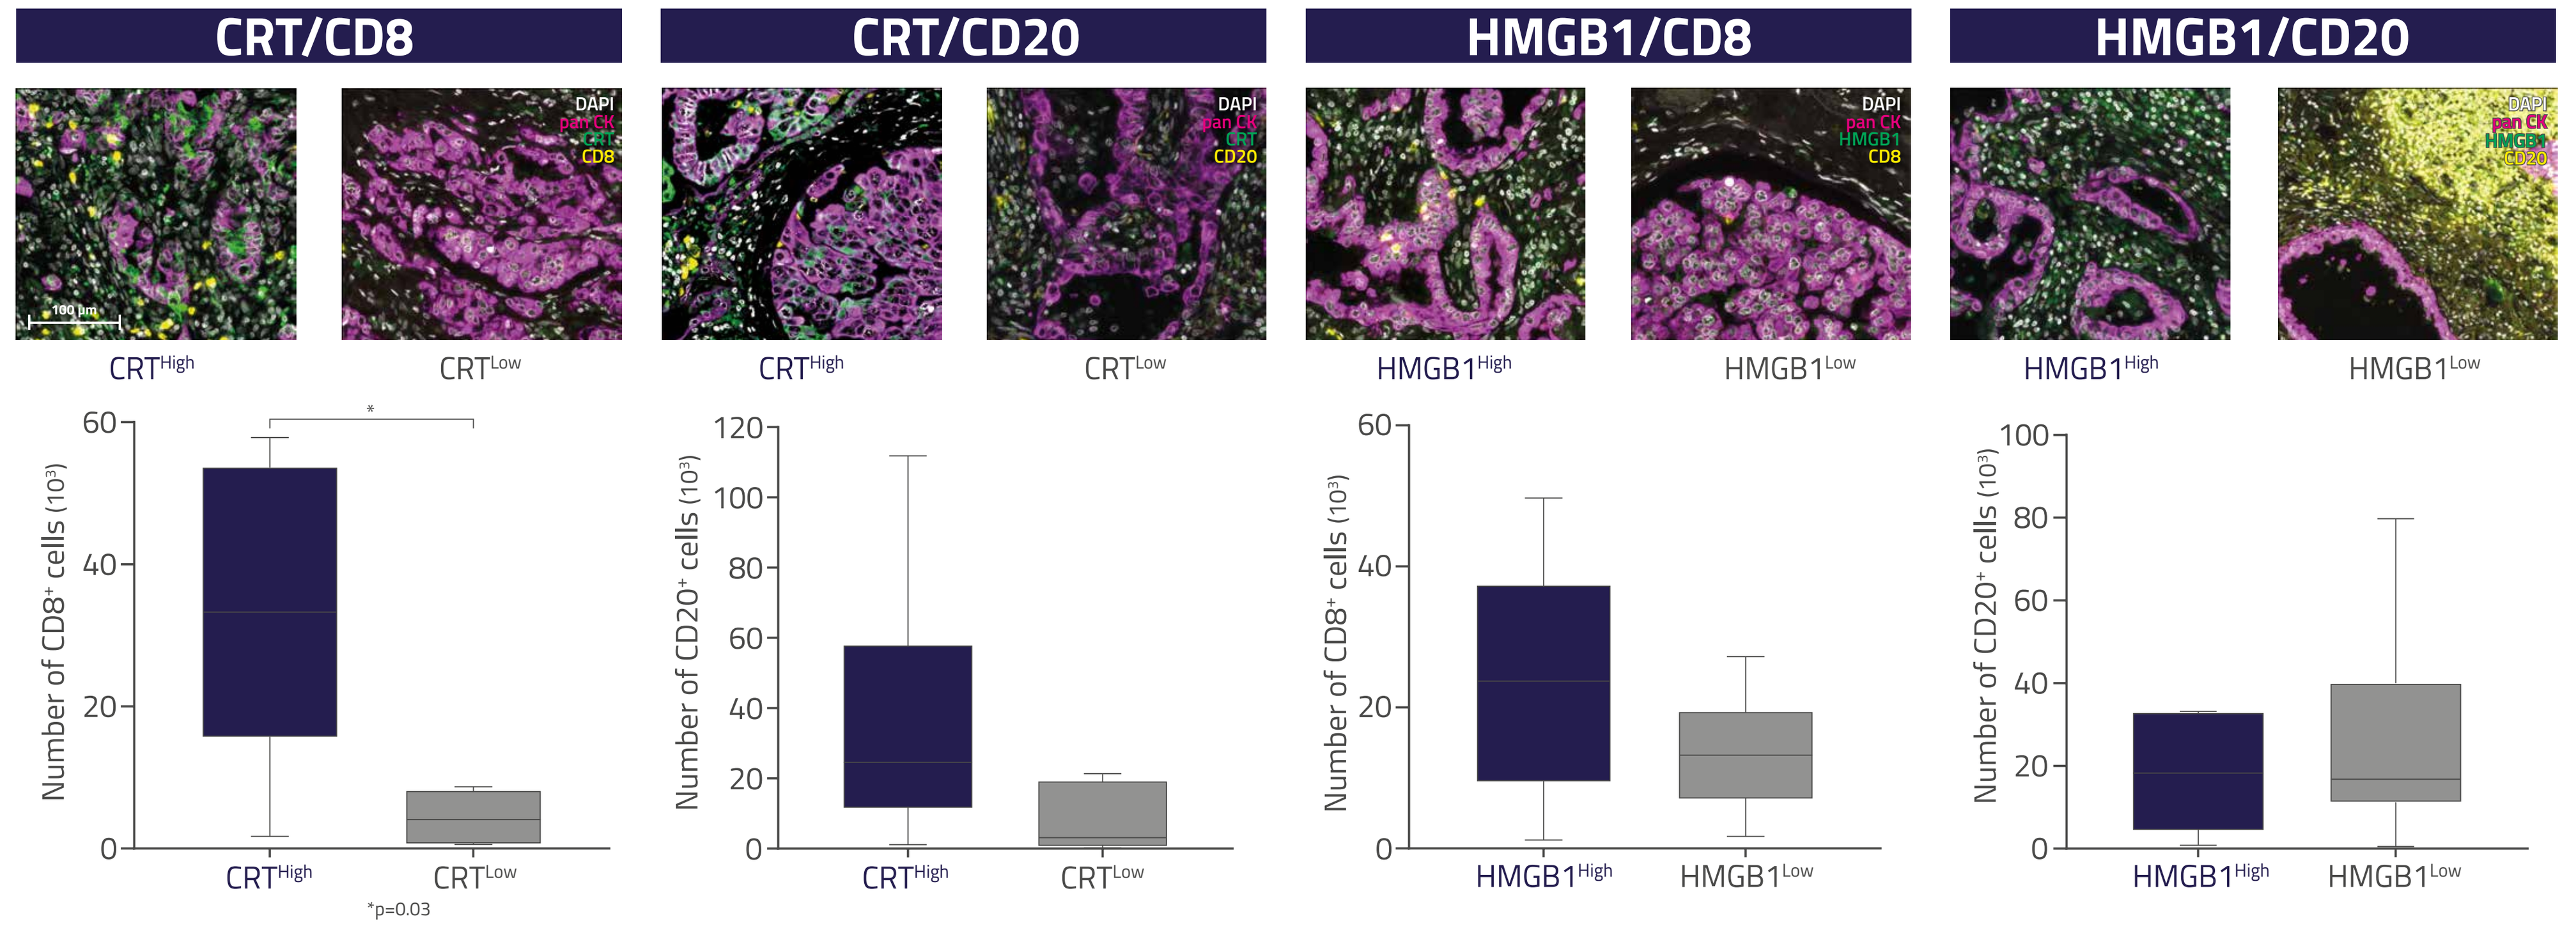

CRT/CD8
CRT/CD20
HMGB1/CD8
HMGB1/CD20
DAPI
pan CK
CRT
CD8
CD20
HMGB1
CRT^{High}
CRT^{Low}
HMGB1^{High}
HMGB1^{Low}
Number of CD8+ cells (10^3)
Number of CD20+ cells (10^3)
*p=0.03

Figure 3. Representative immunofluorescence and quantitation of CRT/CD8, CRT/CD20, HMGB1/CD8, and HMGB1/CD20. Data presented as min, max & median (10th-90th percentile).

- CRT^{High} tumor regions are associated with higher infiltration of CD8+ T cell populations
- HMGB1^{Low} tumor regions are associated with higher infiltration of CD20+ B cell populations

CD8+ cells are most abundant in CRT^{High} tumor regions

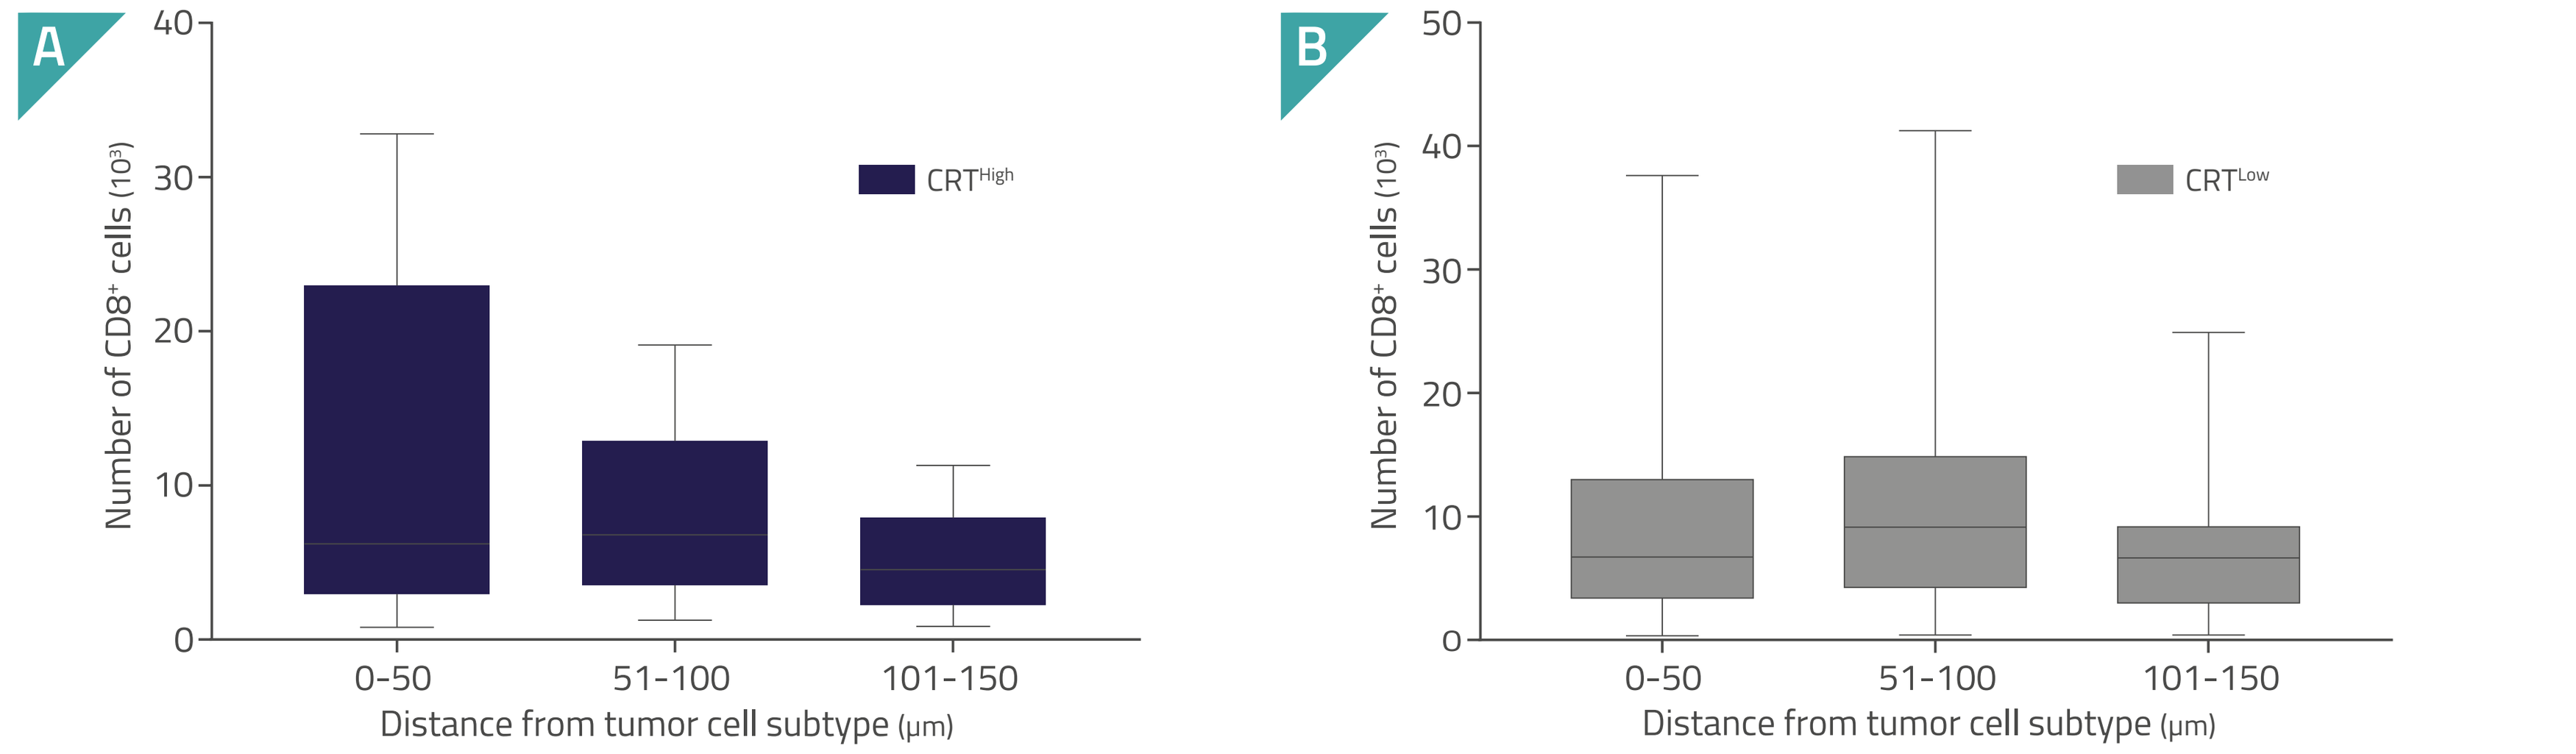

A
B
CRT^{High}
CRT^{Low}
Number of CD8+ cells (10^3)
0-50
51-100
101-150
Distance from tumor cell subtype (µm)

Figure 4. Spatial distribution of CD8+ cytotoxic T cells within (A) CRT^{High} and (B) CRT^{Low} regions. Data presented as min, max & median (10th-90th percentile).

- CD8+ T cells are more abundant in and in closer proximity to CRT^{High} tumor regions

NUC-3373 induces DAMPs in NSCLC cells

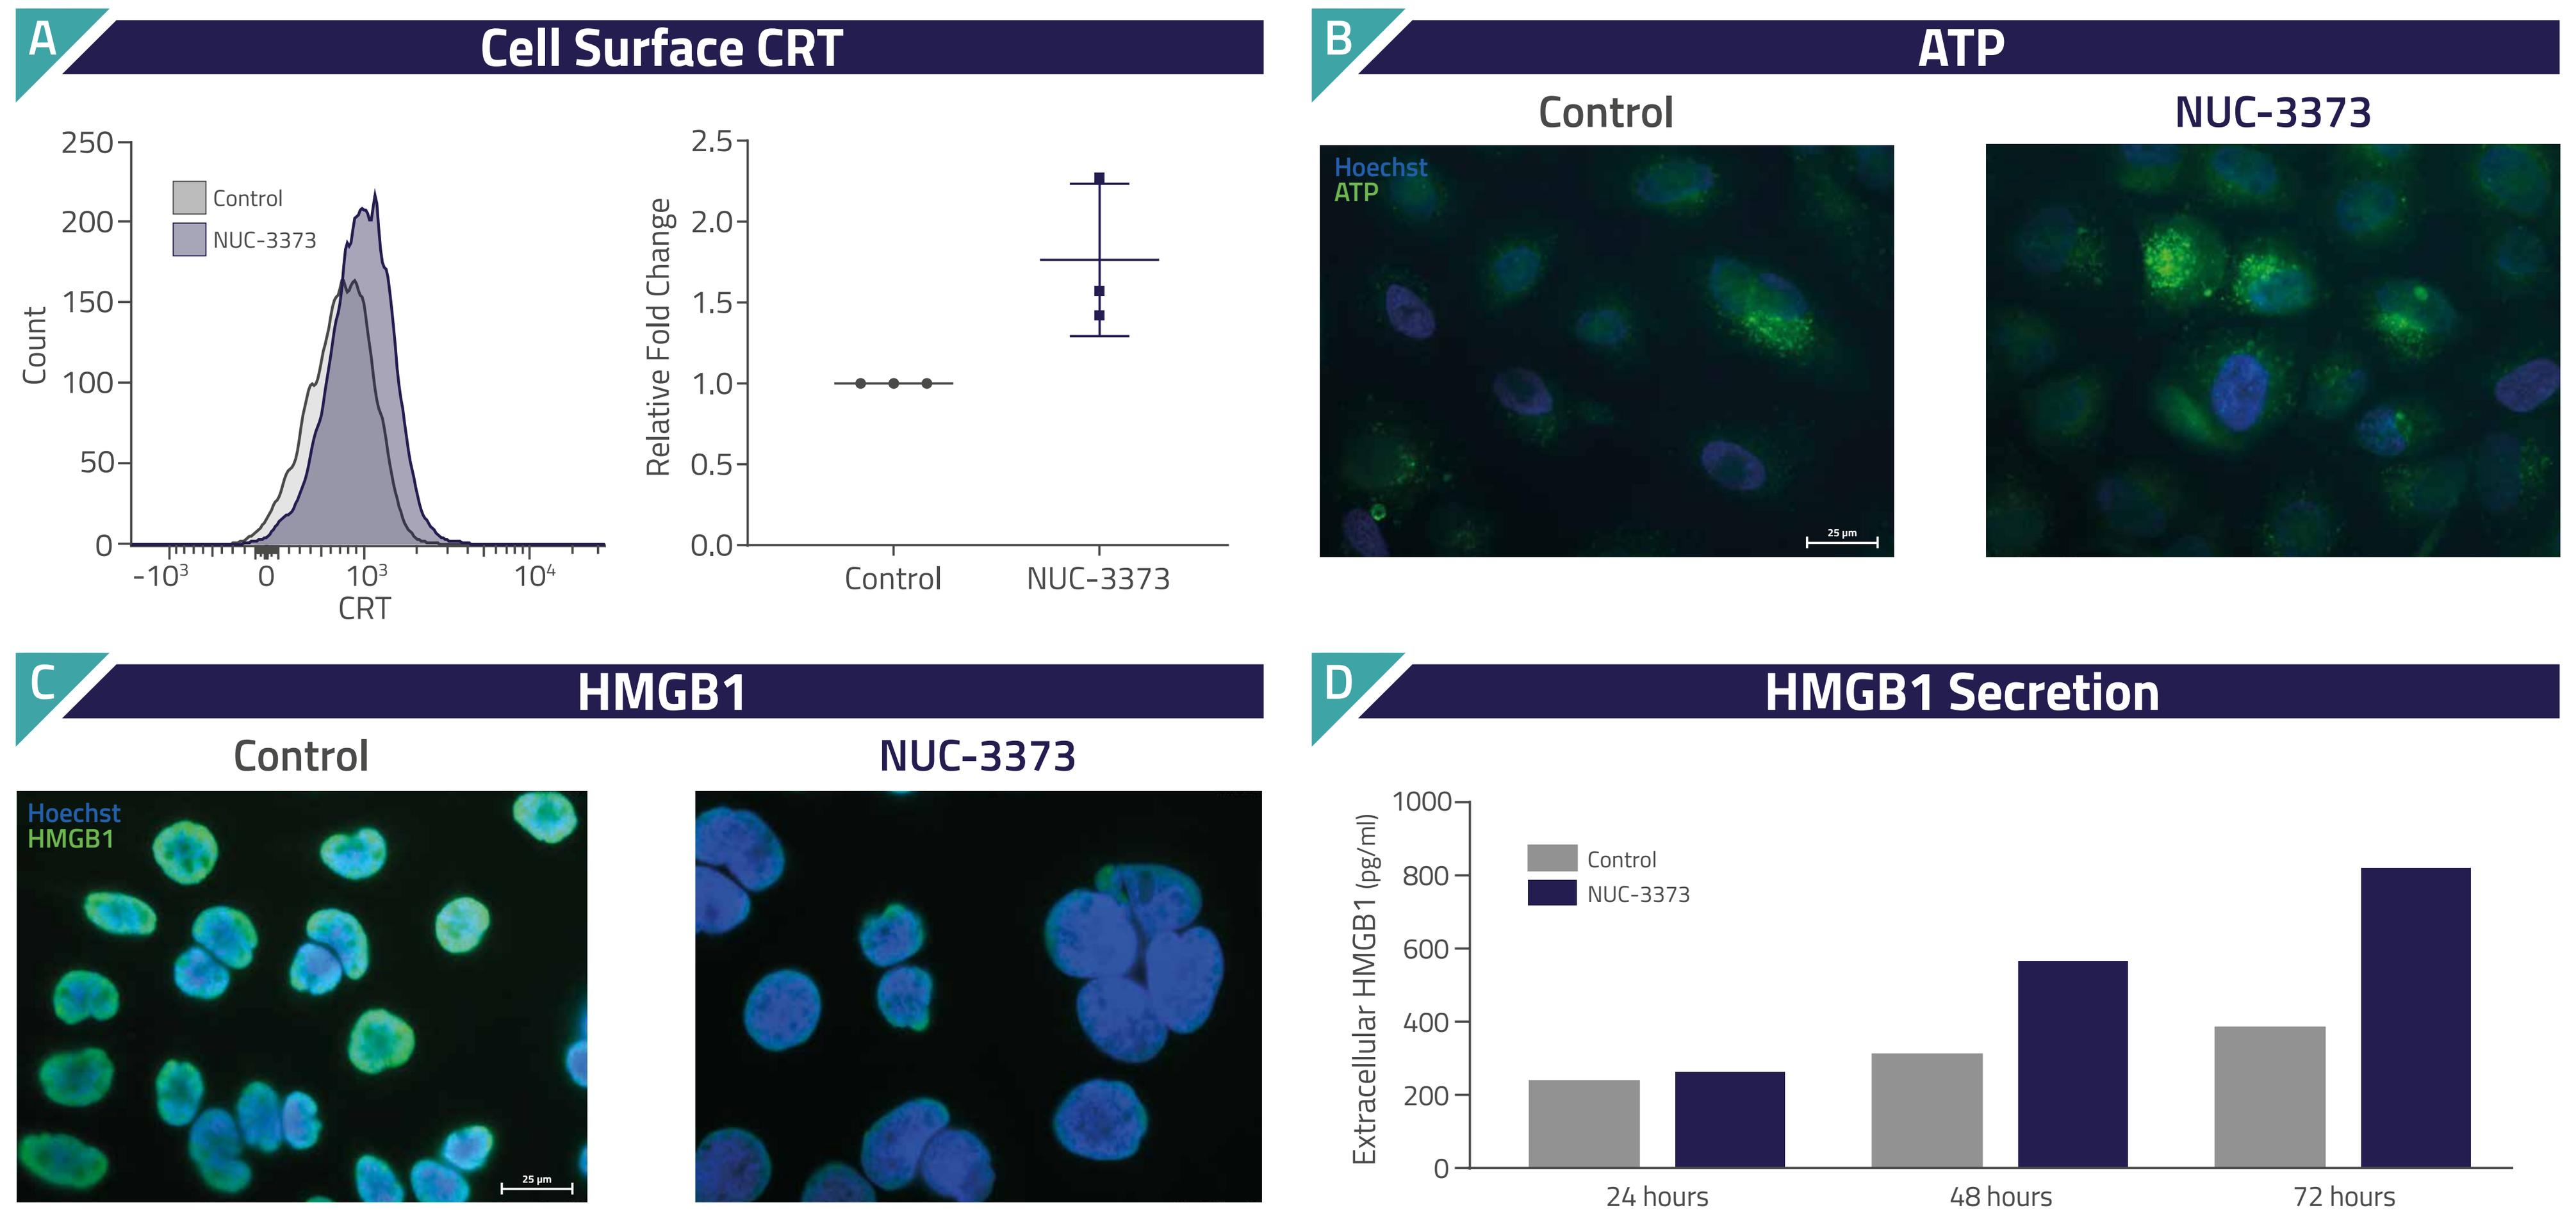

A
Cell Surface CRT
Control
NUC-3373
Count
CRT
Relative Fold Change
B
ATP
Hoechst
ATP
C
HMGB1
Hoechst
HMGB1
D
HMGB1 Secretion
Extracellular HMGB1 (pg/ml)
24 hours
48 hours
72 hours

Figure 5. (A) Cell surface CRT in NSCLC cells treated with NUC-3373 for 24 hours. (B) Extracellular ATP release in NSCLC cells treated with NUC-3373 for 24 hours. (C) Release of nuclear HMBG1 in NSCLC treated with NUC-3373 for 24 hours. (D) Secreted HMGB1 in NSCLC cells treated with NUC-3373 for 24, 48 and 72 hours, n=2.

- NUC-3373 causes the release of DAMPs (CRT, ATP and HMGB1) in NSCLC cells

NUC-3373 increases PD-L1, potentiating T-cell-mediated cytotoxicity when combined with pembrolizumab

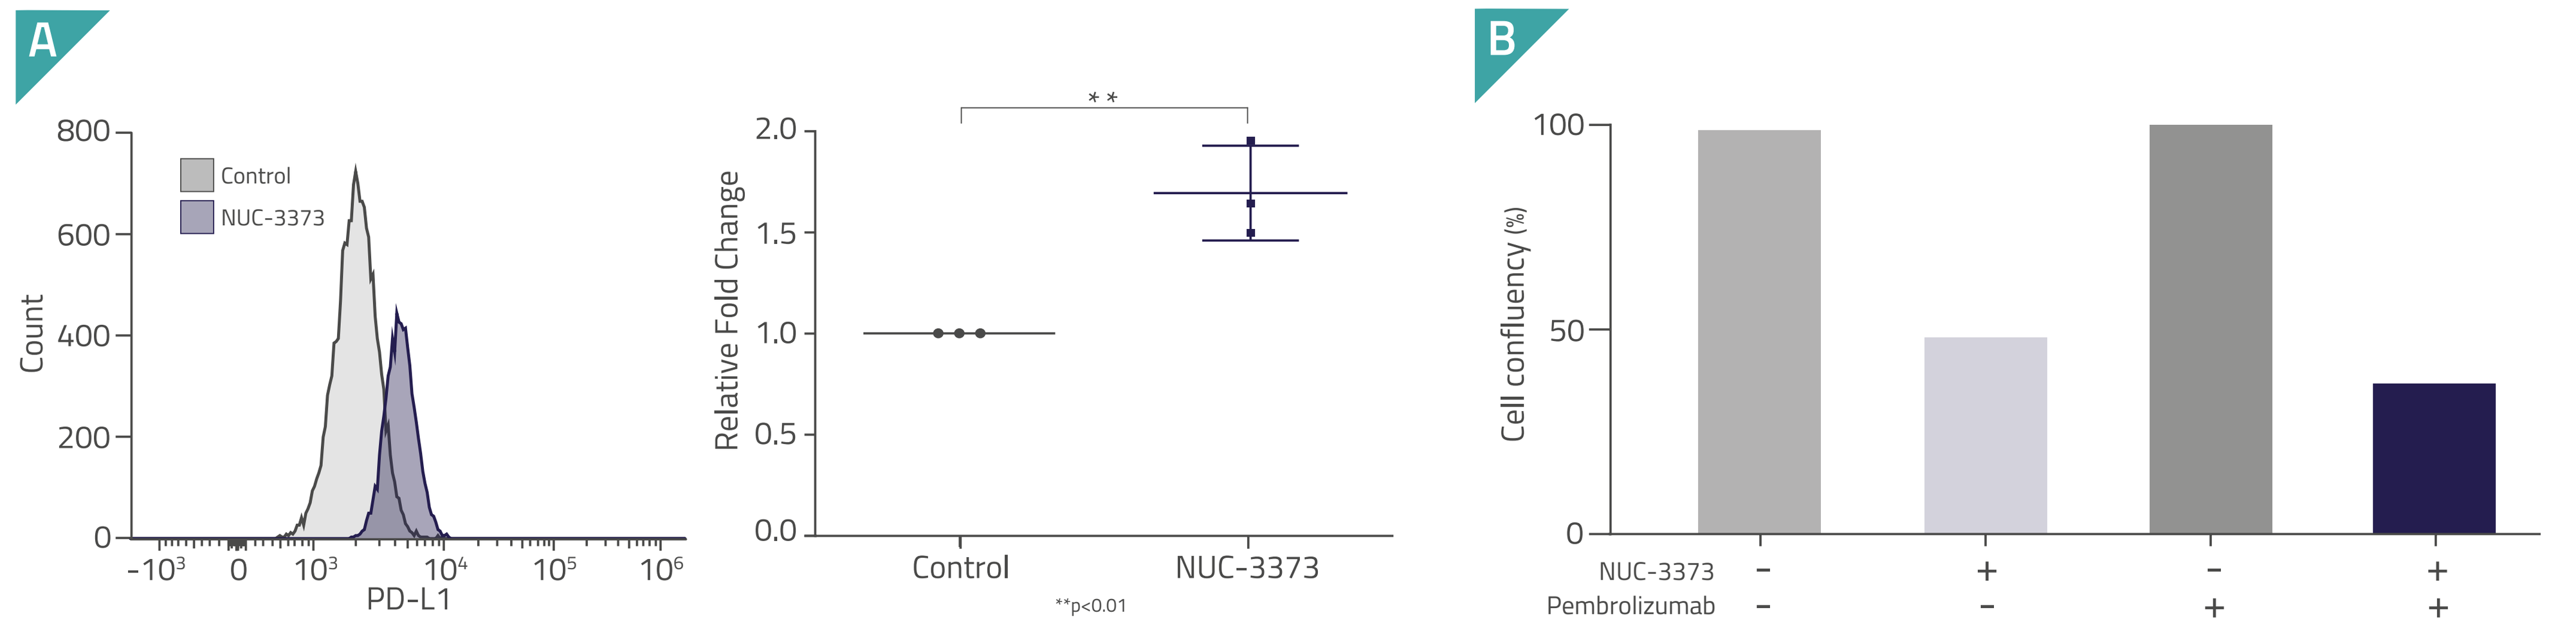

A
Control
NUC-3373
Count
PD-L1
Relative Fold Change
**p<0.01
B
Cell confluency (%)
NUC-3373
Pembrolizumab

Figure 6: (A) PD-L1 surface expression in NSCLC cells treated with NUC-3373 for 24 hours. (B) Confluence of NSCLC cells pre-treated with NUC-3373 and co-cultured with PBMCs for 72 hours (representative data shown).

- NUC-3373 increases cell surface PD-L1 expression in NSCLC cells
- NUC-3373 promotes T-cell mediated cytotoxicity against NSCLC cells, which was increased by pembrolizumab

CONCLUSION

- High tumor immune infiltration predicts improved overall survival in treatment naïve NSCLC patients
- Cell surface DAMP, CRT is associated with high density of CD8+ T cells in treatment naïve NSCLC patients
- NUC-3373 induces DAMPs, PD-L1 and immunogenic cell death in NSCLC cells
- NUC-3373 is an attractive combination partner for immune checkpoint inhibitors

REFERENCES: 1. Ferlay J *et al.*, 2020. *Global Cancer Observatory*: Cancer Today. Lyon, France: International Agency for Research on Cancer. Available from https://gco.iarc.fr/today, accessed [12 Oct 2022] 2. Hernández A *et al.*, 2021. *Cancers*; 13(11): 2821 3. Bruni D G *et al.*, 2020 *Nat Rev Cancer*; 20: 662-680 4. Schulze AB *et al.*, 2020. *J. Thorac. Dis.*; 12(6): 1-19 5. Liu Y *et al.*, 2020. *Theranostics*; 11(11): 5365-5386 6. McGuigan C *et al.*, 2011. *Med Chem*; 27: 7247-7258 7. Vande Voorde J *et al.*, 2011. *Biochem Pharmacol*; 82: 441-452 8. Ciombor K *et al.*, 2020. *J Clin Oncol*; 38 (Suppl 4) 9. Bré J *et al.*, 2022. *Cancer Res*; 82 (Suppl 12): 1835 10. McKissock FG *et al.*, 2020. *Cancer Res*; 80 (Suppl 16): 1848 11. Read OJ *et al.*, 2022. *Cancer Res*; 82 (Suppl 12): 1113

ABBREVIATIONS: ATP: adenosine triphosphate 5-FU: 5-fluorouracil CRT: calreticulin CD8: cluster of differentiation 8 CD20: cluster of differentiation 20 CI: confidence interval DAMP: damage-associated molecular pattern DMSO: dimethyl sulfoxide DNA: deoxyribonucleic acid DPD: dihydropyrimidine dehydrogenase dTMP: deoxythymidine monophosphate dUMP: deoxyuridine monophosphate ER: endoplasmic reticulum FBAL: α-fluoro-ß-alanine FUDR-MP: fluorodeoxyuridine monophosphate FUDR-TP: fluorodeoxyuridine triphosphate FUTP: fluorouridine triphosphate HR: hazard ratio HMGB1: high mobility group box protein 1 ICI: immune checkpoint inhibitor ICD: immunogenic cell death IC_{50}: half-maximal inhibitory concentration NSCLC: non-small cell lung cancer pan CK: pan cytokeratin PBMC: peripheral blood mononuclear cell PD-L1: programmed cell death ligand 1 TME: tumor microenvironment

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.