NUC-3373: A novel pyrimidine nucleotide analogue that overcomes all the main cancer drug resistance mechanisms that limit patient survival

Essam Ghazaly[1], Magdalena Slusarczyk[2], Michaela Serpi[2], Christopher McGuigan[2], David Harrison[3] and Sarah Blagden[4]

1) Barts Cancer Institute, Queen Mary University of London, UK 2) School of Pharmacy & Pharmaceutical Sciences, Cardiff University, UK
3) School of Medicine, University of St Andrews, UK 4) Department of Oncology, University of Oxford, UK

BACKGROUND

NUC-3373

- Novel pyrimidine NucleoTide Analogue
- Preactivated form of the anti-cancer metabolite FdUMP
- ProTide: utilising innovative phosphoramidate chemistry
- Overcomes all the main mechanisms associated with 5-FU, FUDR and capecitabine resistance

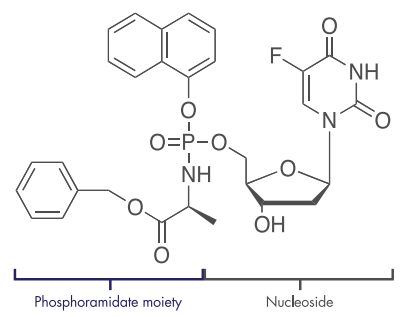
Phosphoramidate moiety
Nucleoside

Figure 1. NUC-3373 structural formula.

5-FU Resistance Mechanisms

Poor drug activation
- Low Thymidine Kinase (TK)
 - TK required for 5-FU activation into active agent FdUMP
 - TK predominantly expressed during S-phase of cell cycle[1]
- High Thymidine Phosphorylase (TP)
 - High levels of TP are associated with adverse survival outcome in colorectal cancer patients receiving 5-FU[2]

Extensive drug degradation
- High Dihydropyrimidine Dehydrogenase (DPD)
 - 80% of 5-FU degraded by DPD
 - Elevated levels of DPD associated with lower overall survival in colorectal cancer patients receiving capecitabine[3]

Limited cellular uptake
- Low human Equilibrative Nucleoside Transporter 1 (hENT1)
 - Nucleobase and nucleoside transporters required for 5-FU / FUDR cellular uptake

Weak target inhibition
- High Thymidylate Synthase (TS), a target of 5-FU
 - Elevated levels of TS are associated with poor survival in colorectal cancer
 - High TS is a predictive factor for poor response to 5-FU[4]

Mycoplasma infection
- Approximately 55% of colorectal cancers are infected by *Mycoplasma*[5]
- In cancer cell lines, *Mycoplasma* infection decreases 5-FU activity by up to 100x[6]

MODE OF ACTION / METABOLISM

NUC-3373 overcomes all the main 5-FU resistance pathways

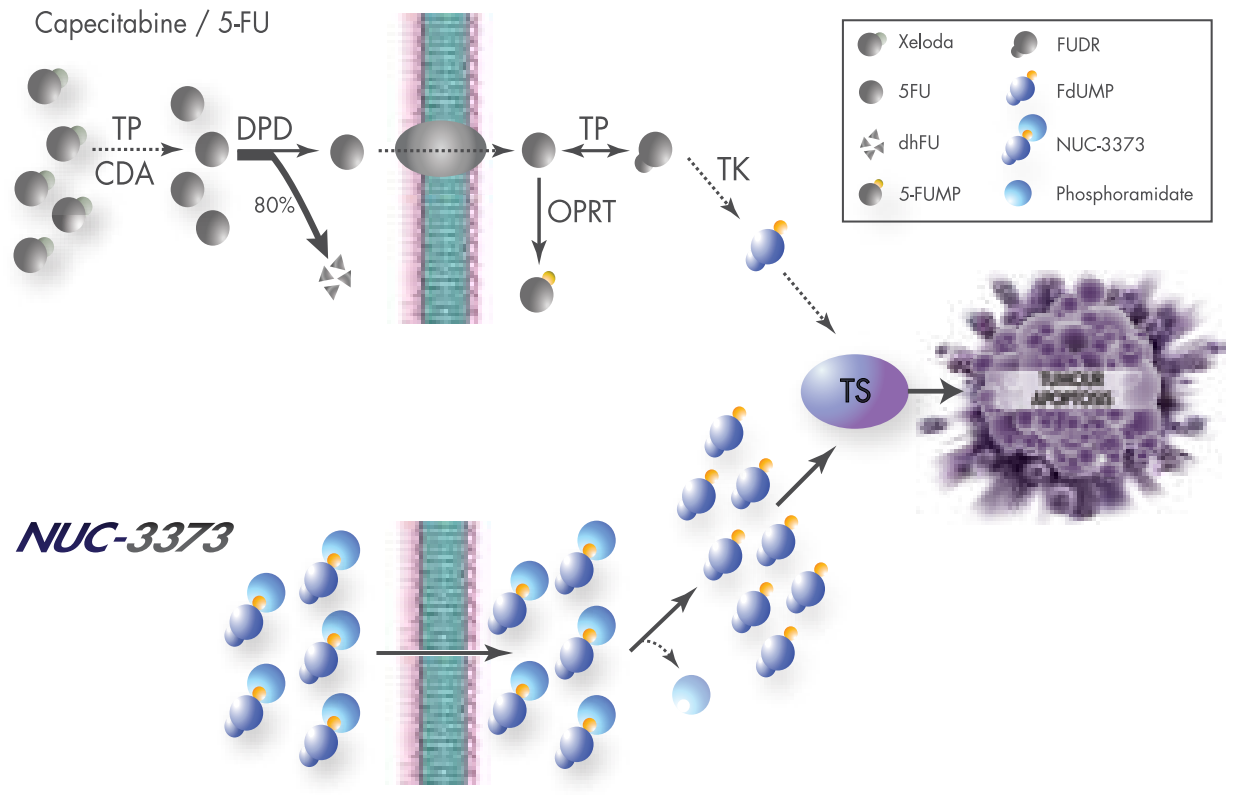
Capecitabine / 5-FU
TP
CDA
DPD
80%
TP
OPRT
TK
TS
Xeloda
5FU
dhFU
5-FUMP
FUDR
FdUMP
NUC-3373
Phosphoramidate
NUC-3373

Top: 5-FU, FUDR and capecitabine require active uptake and multi-step metabolism for conversion into the active anti-cancer agent FdUMP.

Bottom: NUC-3373 is a preactivated form of FdUMP bearing a phosphoramidate moiety (ProTide). NUC-3373 enters the cell independent of nucleoside transporters where the moiety is cleaved to release high levels of active FdUMP intracellularly which directly inhibits Thymidylate Synthase (TS) and induces apoptosis.

METHODS

Cytotoxicity

- Establish EC_{50} values in multiple cancer cells including 5-FU resistant lines
- Measure efficacy in resistance conditions using TK^- and $hENT1^-$ cells
- Compare activity in *Mycoplasma* infected cancer cells

Metabolism

- Quantify intracellular levels of the active moiety FdUMP in colorectal cancer cells
- Evaluate the effect of Dihydropyrimidine Dehydrogenase (DPD) on drug metabolism

Xenograft

- Validate activity *in vivo* utilising human colorectal cancer cells (HT29)

Toxicology

- Conduct toxicology studies to establish safety profile

RESULTS

NUC-3373 is a potent anti-cancer agent

- NUC-3373 demonstrates up to 330x greater activity than 5-FU across a broad range of 5-FU sensitive and resistant cancer cell lines

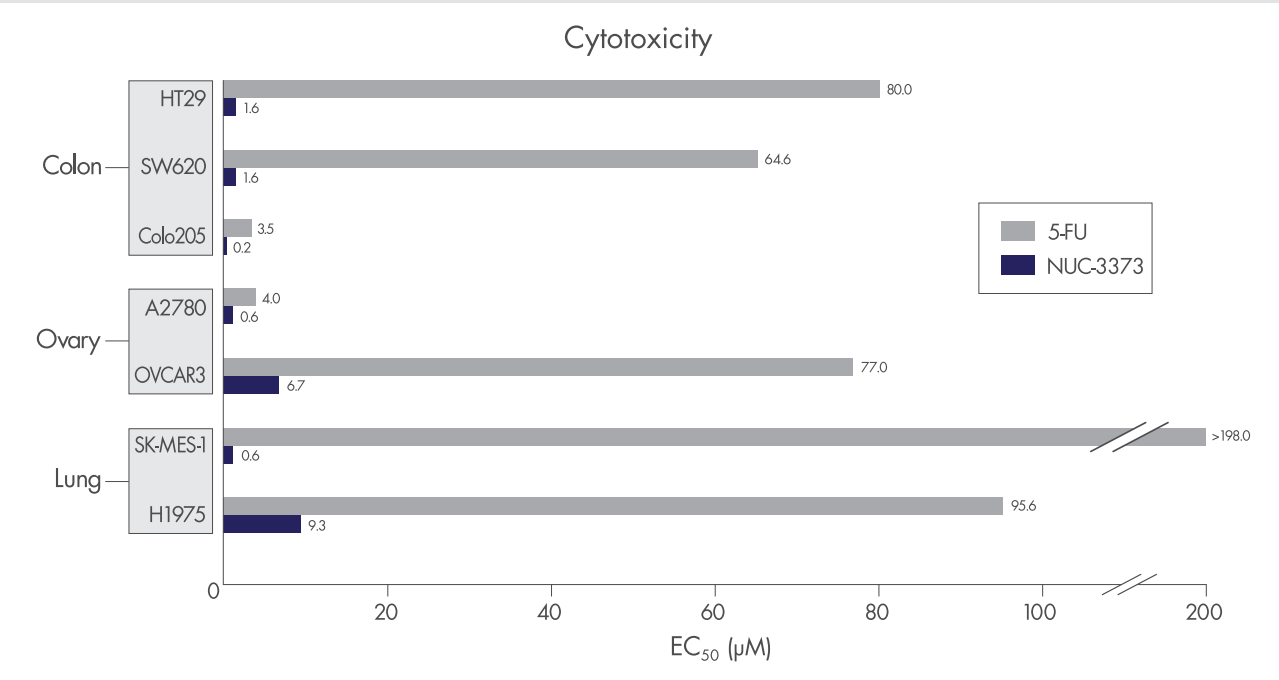
Cytotoxicity
Colon
HT29
SW620
Colo205
Ovary
A2780
OVCAR3
Lung
SK-MES-1
H1975
80.0
1.6
64.6
1.6
3.5
0.2
4.0
0.6
77.0
6.7
>198.0
0.6
95.6
9.3
5-FU
NUC-3373
0
20
40
60
80
100
200
EC50 (μM)

Figure 2. Comparative anti-proliferative effect of NUC-3373 and 5-FU in colorectal, ovarian and lung cancer cell lines. EC_{50} values standardised for molecular weight.

NUC-3373 overcomes all the main cancer resistance mechanisms

- Thymidine Kinase (TK)
 - FUDR activity is significantly impaired in TK deficient cancer cells whilst that of NUC-3373 is unchanged
- Thymidine Phosphorylase (TP)
 - NUC-3373 is resistant to TP degradation

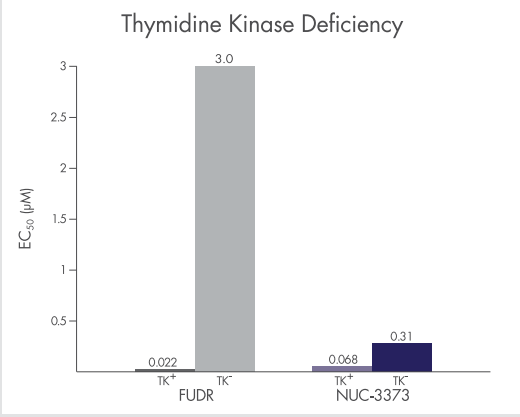
Thymidine Kinase Deficiency
EC50 (μM)
3.0
0.022
0.068
0.31
TK+
TK-
FUDR
NUC-3373

Figure 3. NUC-3373 activity independent of TK while FUDR loses activity in TK-deficient CEM human leukaemia cancer cell line.

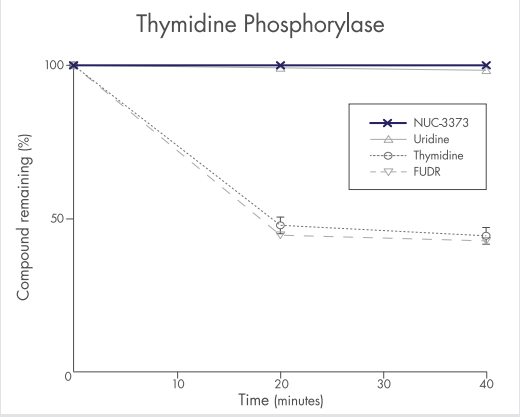
Thymidine Phosphorylase
Compound remaining (%)
NUC-3373
Uridine
Thymidine
FUDR
Time (minutes)

Figure 4. TP purified from human erythrocytes was incubated with NUC-3373 and FUDR. NUC-3373 is resistant to TP degradation while FUDR is significantly broken down by TP.

- Nucleoside Transporter (hENT1)
 - FUDR activity is reduced by 63x in hENT1-deficient cancer cells (CEM) while NUC-3373 maintains effective anti-proliferative activity
 - The cellular uptake of NUC-3373 is independent of nucleoside transporters

NUC-3373 is resistant to DPD degradation

- 5-FU levels are significantly elevated in the presence of a DPD inhibitor (gimeracil) while NUC-3373 levels remain unaffected

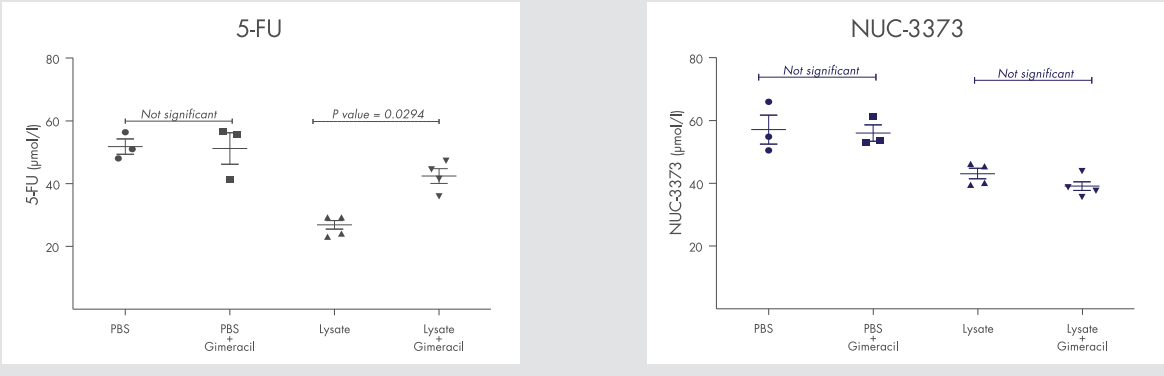
5-FU
5-FU (μmol/l)
Not significant
P value = 0.0294
PBS
PBS + Gimeracil
Lysate
Lysate + Gimeracil
NUC-3373
NUC-3373 (μmol/l)
Not significant
Not significant

Figure 5. NUC-3373 and 5-FU levels in combined SW620, HCT-116 and HT29 colorectal cancer cell lysates +/- gimeracil (DPD inhibitor).

NUC-3373 achieves high intracellular levels of active agent FdUMP

- FdUMP is the main active anti-cancer metabolite of 5-FU / FUDR
- NUC-3373 generates FdUMP intracellular levels 363x higher than 5-FU
- FdUMP levels generated by NUC-3373 remain high in all the key cancer resistance-like conditions (overexpression of TS, DPD, TP, OPRT, CDA)

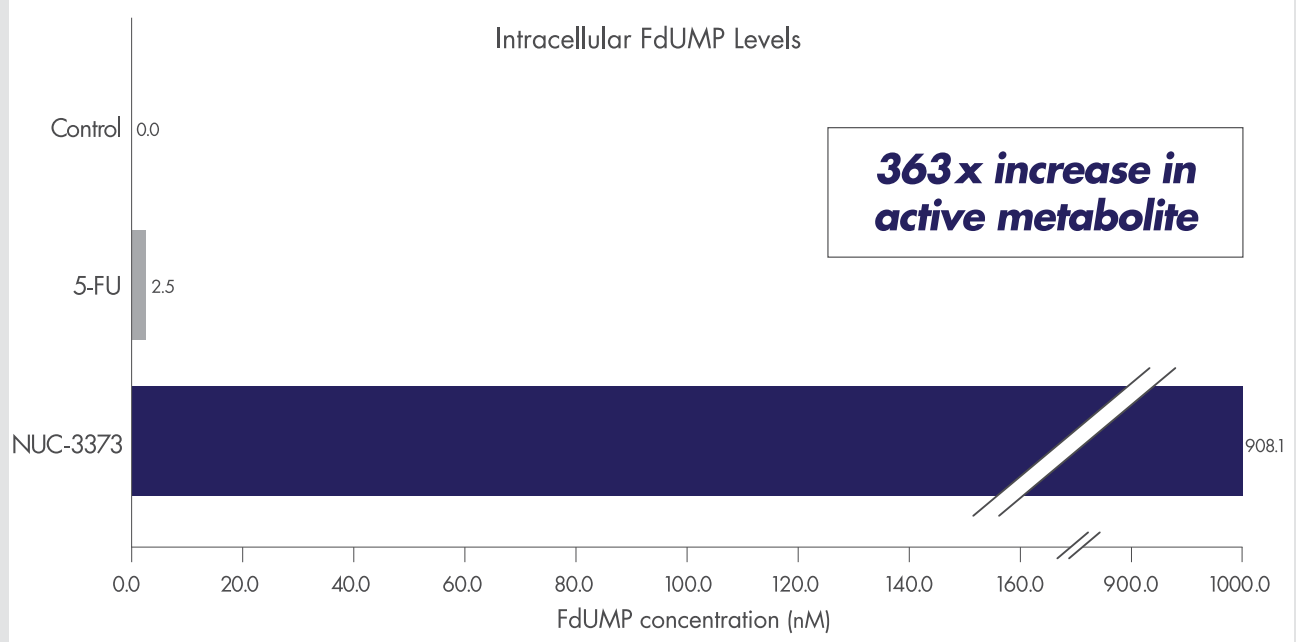
Intracellular FdUMP Levels
Control
0.0
5-FU
2.5
NUC-3373
908.1
363x increase in active metabolite
FdUMP concentration (nM)

Figure 6. Intracellular levels of FdUMP generated by NUC-3373 and 5-FU in human colorectal cancer cell line (HT29).

NUC-3373 activity is unaffected by *Mycoplasma* infection

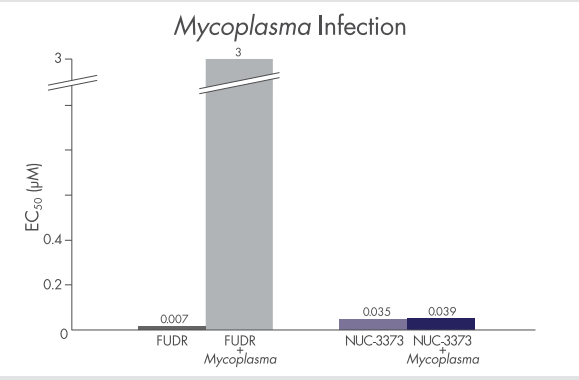
Mycoplasma Infection
EC50 (μM)
0.007
3
0.035
0.039
FUDR
FUDR + Mycoplasma
NUC-3373
NUC-3373 + Mycoplasma

- Unlike FUDR, NUC-3373 maintains activity in the presence of *Mycoplasma*

Figure 7. Anti-proliferative effect of NUC-3373 and FUDR in L1210 leukaemic cancer cell line chronically infected by Mycoplasma hyorhinis.

NUC-3373 demonstrates superior inhibition of tumour growth *in vivo*

- NUC-3373 achieves significantly greater reduction in tumour weight and volume than 5-FU in HT29 colorectal xenografts
- NUC-3373 significantly prolongs tumour volume doubling time compared to control (vehicle)

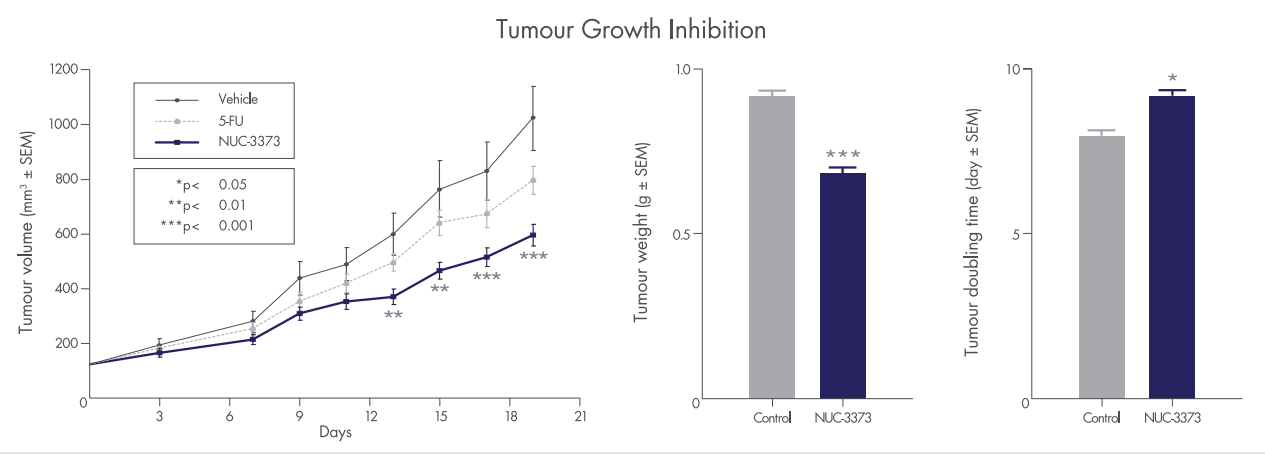
Tumour Growth Inhibition
Vehicle
5-FU
NUC-3373
*p< 0.05
**p< 0.01
***p< 0.001
Tumour volume (mm³ ± SEM)
Days
Tumour weight (g ± SEM)
Tumour doubling time (day ± SEM)
Control
NUC-3373

Figure 8. NUC-3373 and 5-FU effect on HT29 human colorectal cancer xenografts. Control = vehicle.

NUC-3373 demonstrates a favourable toxicology profile

- In formal toxicology studies NUC-3373 is significantly better tolerated than 5-FU
- The main toxicities associated with 5-FU after a single dose are not observed with NUC-3373 after single or repeat dosing using equivalent or higher levels of drug
- NUC-3373 is rapidly distributed into tissues following IV bolus administration
- Plasma AUC ratios show low conversion of NUC-3373 into FUDR and dhFU (metabolite : NUC-3373 of 0.01 and 0.03 respectively)

Table 1. AUC and C_{max} of NUC-3373, FUDR and dhFU in plasma.

Analyte	Mean $AUC_{(0-t)}$	Mean C_{max}
NUC-3373	1745.0 ng.h/ml	5295.0 ng/ml
dhFU	38.5 ng.h/ml	38.6 ng/ml
FUDR	18.7 ng.h/ml	24.1 ng/ml

Day 28 values dosed at 8mg/kg/day for 5 consecutive days

CONCLUSIONS

- NUC-3373 is a novel pyrimidine nucleotide analogue that overcomes all the main cancer resistance mechanisms associated with 5-FU, FUDR and capecitabine.
- Significantly greater anti-proliferative activity observed with NUC-3373 than 5-FU across a broad range of sensitive and resistant cancer cells including *Mycoplasma* infected cells.
- NUC-3373 generates 363x higher intracellular levels of the active agent, FdUMP, than 5-FU.
- NUC-3373 significantly decreases tumour weight and volume compared to 5-FU *in vivo* and is significantly better tolerated in toxicology studies.
- First In Human Phase I study of NUC-3373 will commence in Q4 2015.
- Results demonstrate that NUC-3373 has the potential to replace 5-FU as the standard of care for colorectal cancer and other solid tumours.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from AACR-NCI-EORTC® and the author of this poster.

References: [1] Huang et al., 1994 [2] Takebayashi et al., 1996 [3] Koopman et al., 2009 [4] Peters et al., 1995 [5] Huang et al., 2001 [6] Jette et al., 2008

AACR-NCI-EORTC